131P - Translating a novel autotaxin inhibitor from preclinical proof of concept in pancreatic cancer to a biomarker response in human subjects

S Milleri[1], MA Deken[2], R Medhi[3], A Carruthers[3], A Tagliavini[4], A Nizzardo[4], M Pergher[4], K Niewola-Staszkowska[2], A Cheasty[3], A Fraser[3], P Shah[3], L van der Veen[2], L Ziviani[1], M Lahn[2] & Z Johnson[2]

[1]Centro Ricerche Cliniche di Verona srl Verona, Italy, [2]iOnctura SA, Geneva, Switzerland, [3]Cancer Research UK Therapeutic Discovery Laboratories, Cambridge, UK, [4]Evotec, Verona, Italy

BACKGROUND

Autotaxin (ATX) is a secreted glycoprotein that hydrolyzes lysophosphatidylcholine (LPC) to lysophosphatidic acid (LPA). LPA signaling directly modulates tumor cell function and contributes to the development of the fibrotic tumor microenvironment, resulting in reduced host immunity and impaired response to therapy. The expression of ATX and LPA is elevated in the plasma of pancreatic cancer (PC) patients and correlates with soluble CA19-9. IOA-289 is a potent and orally available autotaxin inhibitor which is being developed as a novel treatment for pancreatic cancer and other highly fibrotic tumors.

METHODS

PK/PD studies were performed following a single oral dose of IOA-289 in mice, and plasma LPA was used as a PD biomarker (fig. 1)
Plasma concentrations of IOA-289 and LPA were measured following a single oral dose of IOA-289 in a healthy volunteer study (fig. 2)
Analysis of ATX (gene name ENPP2) expression in various cancers and sample types using TCGA and GTEX data indicated that ENPP2 expression is increased in Pancreatic adenocarcinoma (PDAC) compared to normal tissue (fig. 3A). The direct, anti-proliferative activity of IOA-289 on MiaPaCa2 cells was studied in a similar assay (fig. 3B). In vivo efficacy was studied in an orthotopic mouse model of pancreatic cancer (fig. 3C). The effect of cancer associated fibroblasts (CAFs) on proliferation of the human pancreatic cancer cell line, MiaPaCa2 was studied in vitro (fig. 3D).

RESULTS

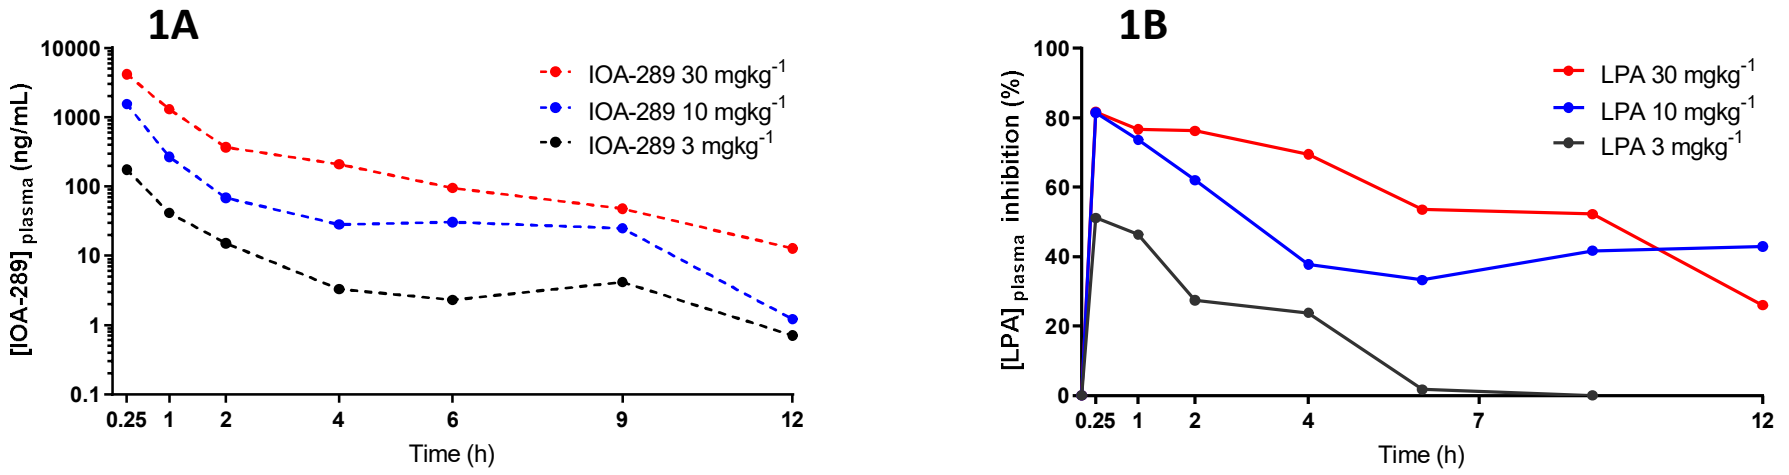
1A
[IOA-289] plasma (ng/mL)
IOA-289 30 mgkg-1
IOA-289 10 mgkg-1
IOA-289 3 mgkg-1
Time (h)
1B
[LPA] plasma inhibition (%)
LPA 30 mgkg-1
LPA 10 mgkg-1
LPA 3 mgkg-1
Time (h)

Figure 1 Mouse PK and PD: Male CD1 mice dosed with 3, 10 or 30 mg/kg of IOA-289 p.o. showed dose-dependent reduction of circulating LPA C18:2 with an ED50 value at 1 h post-dose of around 3 mg/kg (fig. 1A and 1B).

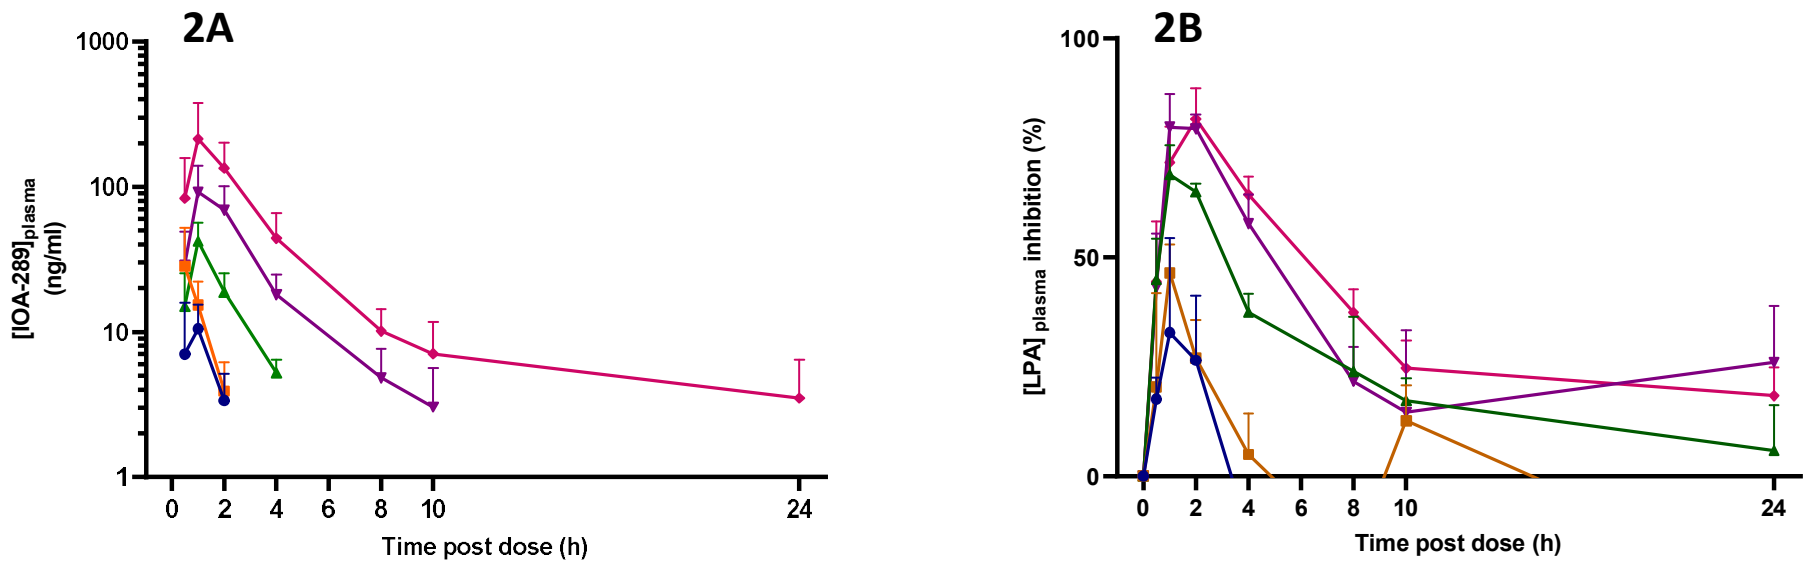
2A
[IOA-289]plasma (ng/ml)
Time post dose (h)
2B
[LPA] plasma inhibition (%)
Time post dose (h)

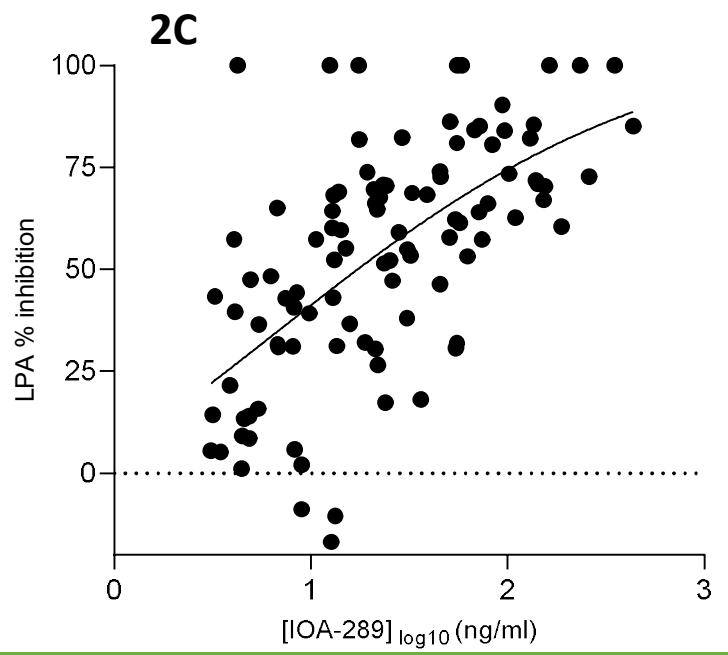
2C
LPA % inhibition
[IOA-289] log10 (ng/ml)

Figure 2 Randomized, double-blind, placebo-controlled study of single ascending oral doses of IOA-289: Healthy male volunteers were administered a single dose of IOA-289 in a capsule formulation (6 subjects per cohort). Plasma exposure was measured up to 24 h post dose (fig. 2A). LPA C18:2 in plasma was measured at baseline and up to 24 h post dose, data are % inhibition of baseline levels (fig. 2B). Using the PK and PD data, the IC_{50} of IOA-289 was calculated as 15 ng/mL in human plasma (fig. 2C)

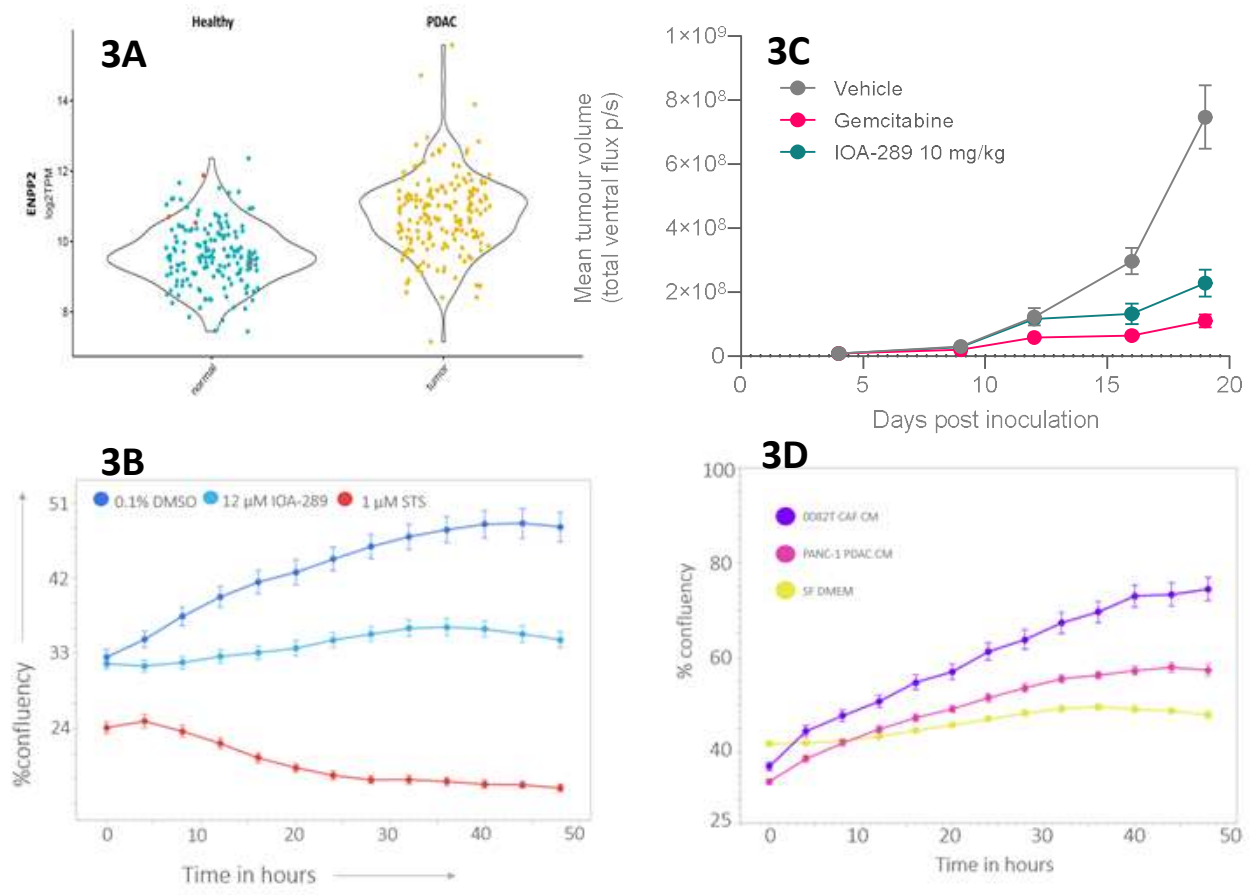
3A
Healthy
PDAC
ENPP2 log2TPM
normal
tumor
3C
Vehicle
Gemcitabine
IOA-289 10 mg/kg
Mean tumour volume (total ventral flux p/s)
Days post inoculation
3B
0.1% DMSO
12 µM IOA-289
1 µM STS
%confluency
Time in hours
3D
0082T CAF CM
PANC-1 PDAC CM
SF DMEM
% confluency
Time in hours

Figure 3 ATX drives PDAC growth: Pancreatic adenocarcinoma (PDAC) has high ENNP2 expression compared to normal tissue (fig. 3A). The growth of MIA PaCa-2 pancreatic cancer cells is inhibited by IOA-289 (fig. 3B). IOA-289 reduced tumor burden *in vivo* in a mouse model of pancreatic cancer (fig. 3C). An increase in MIA PaCa-2 proliferation is observed in the presence of conditioned media generated from the 0082T pancreatic CAF cell line (fig. 3D).

CONCLUSION

- IOA-289 is a potent, orally available ATX inhibitor
- IOA-289 PK/PD relationship was translated from mouse to human with a plasma IC50 of 15 ng/mL
- ATX is elevated in PDAC and directly impacts cancer cell line growth *in vitro*
- IOA-289 reduced tumor burden in a mouse model of PC
- Cancer associated fibroblasts stimulate PDAC cell growth *in vitro* and further studies will investigate the role of ATX in the CAF/tumor interface
- IOA-289 is in clinical development for the treatment of solid tumors burdened with a high degree of fibrosis

ESMO | IMMUNO-ONCOLOGY CONGRESS | Dec 8-11 2021 Presenter´s email: stefano.milleri@crc.vr.it Corresponding email: z.johnson@ionctura.com NCT05027568 - Sponsored by iOnctura SA